A phase I/IIa study of the human anti-CD38 antibody MOR202 (MOR03087) in relapsed or refractory multiple myeloma (rrMM)

Marc S. Raab[1], Hartmut Goldschmidt[1], Hermine Agis[2], Igor W. Blau[3], Hermann Einsele[4], Monika Engelhardt[5], Barbara Ferstl[6], Martin Gramatzki[7], Christoph Röllig[8], Katja Weisel[9], Pia Klöpfer[10], Dominika Weinelt[10], Stefan Härtle[10], Christian Peschel[11]

[1]University Hospital Heidelberg, Heidelberg, Germany; [2]University Hospital of Internal Medicine – AKH Wien, Vienna, Austria; [3]Charité Campus Benjamin Franklin, Berlin, Germany; [4]University Hospital Würzburg, Würzburg, Germany; [5]Medical University Hospital, Freiburg, Germany; [6]Friedrich-Alexander-University Erlangen-Nürnberg, Erlangen, Germany; [7]University Hospital Schleswig-Holstein Division of Stem Cell Transplantation and Immunotherapy, Kiel, Germany; [8]University Hospital Carl Gustav Carus, Dresden, Germany; [9]University Hospital of Tübingen, Tübingen, Germany; [10]MorphoSys AG, Martinsried, Germany; [11]Technical University of Munich, Munich, Germany

ASCO Annual Meeting, May 29 – June 2, 2015: abstract no. 8574

Introduction

- CD38 is a 45 kDa type II transmembrane glycoprotein widely expressed in many hematologic malignancies including multiple myeloma (MM).[1–5]
- MOR202 is a HuCAL-derived human immunoglobulin G1 anti-CD38 antibody that has demonstrated high *in vitro* and *in vivo* efficacy in preclinical models of MM.
- The main mode of action for MOR202-induced lysis of MM cells comprises antibody-dependent cellular cytotoxicity (ADCC) and phagocytosis (ADCP).[6–7]

Figure 1. MOR202 mode of action

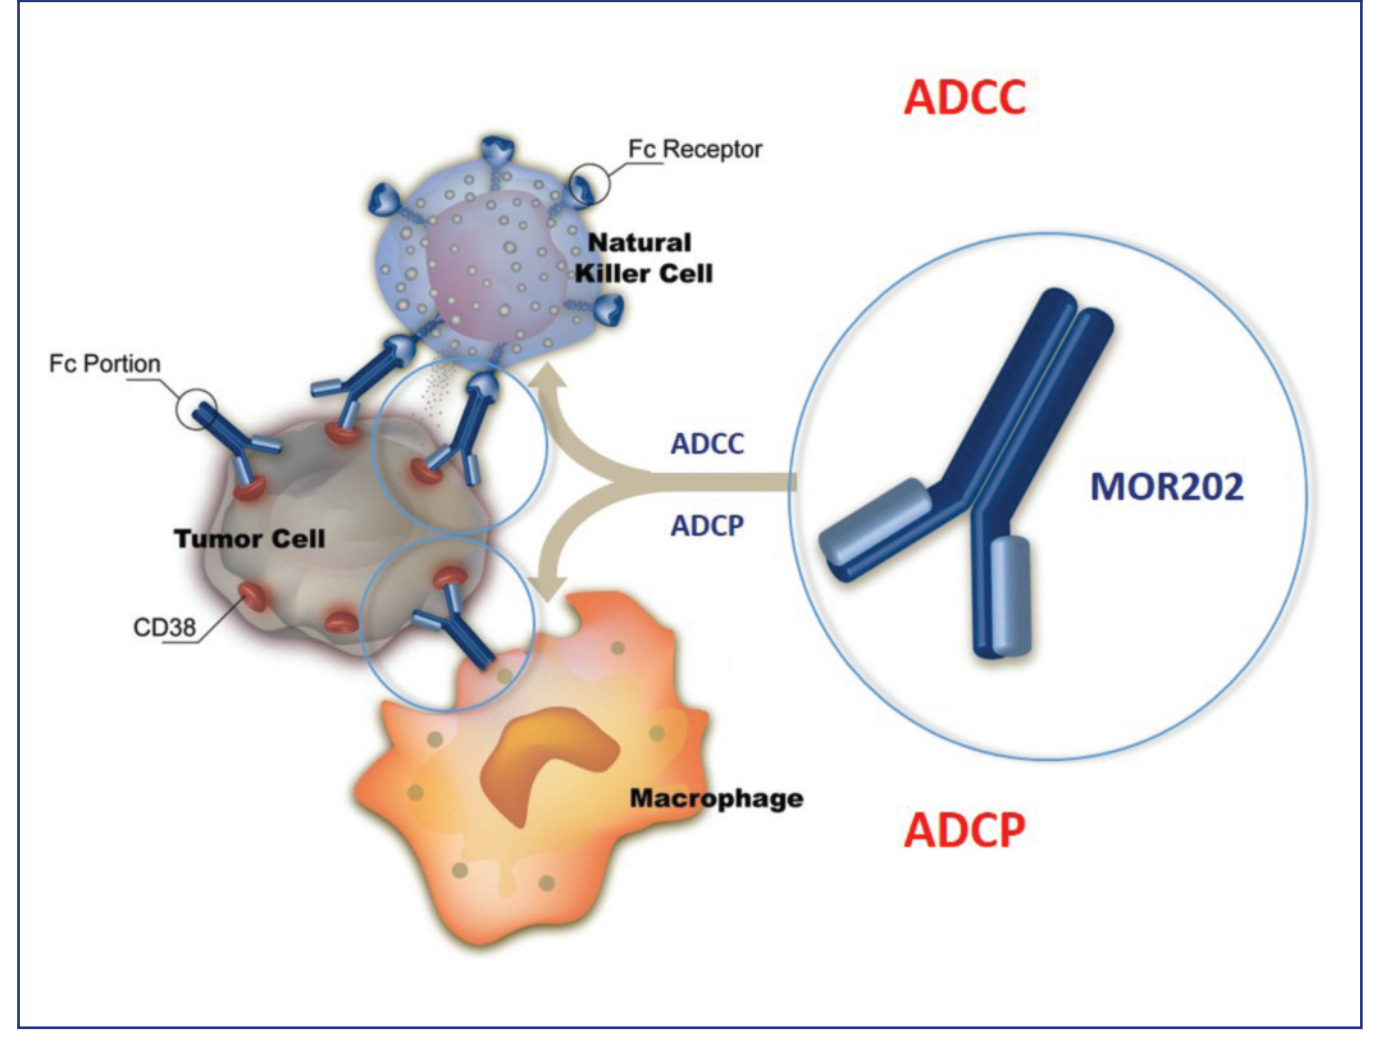
ADCC
Fc Receptor
Natural Killer Cell
Fc Portion
ADCC
ADCP
MOR202
Tumor Cell
CD38
Macrophage
ADCP

ADCC, antibody-dependent cellular cytotoxicity; ADCP, antibody-dependent cellular phagocytosis.

Study objectives

- To evaluate MOR202 in adult patients with relapsed or refractory MM (rrMM).
- **Primary outcome measures:**
 - Maximum tolerated dose (MTD) and/or recommended dose/schedule of MOR202 as monotherapy with and without dexamethasone (DEX), and in combination with pomalidomide (POM)/DEX and lenalidomide (LEN)/DEX (see Figure 2 footnote)
 - Safety: Incidence and severity of adverse events (AEs)
 - Immunogenicity of MOR202.
- **Secondary outcome measures:**
 - Pharmacokinetics (PK) of MOR202 without and with POM/DEX and LEN/DEX
 - Overall response rate, duration of response, time-to-progression, and progression-free survival.

Overall study design

- This is a phase I/IIa open-label, multicenter, dose-escalation study (3 + 3 design; Figure 2).

Figure 2. Study design

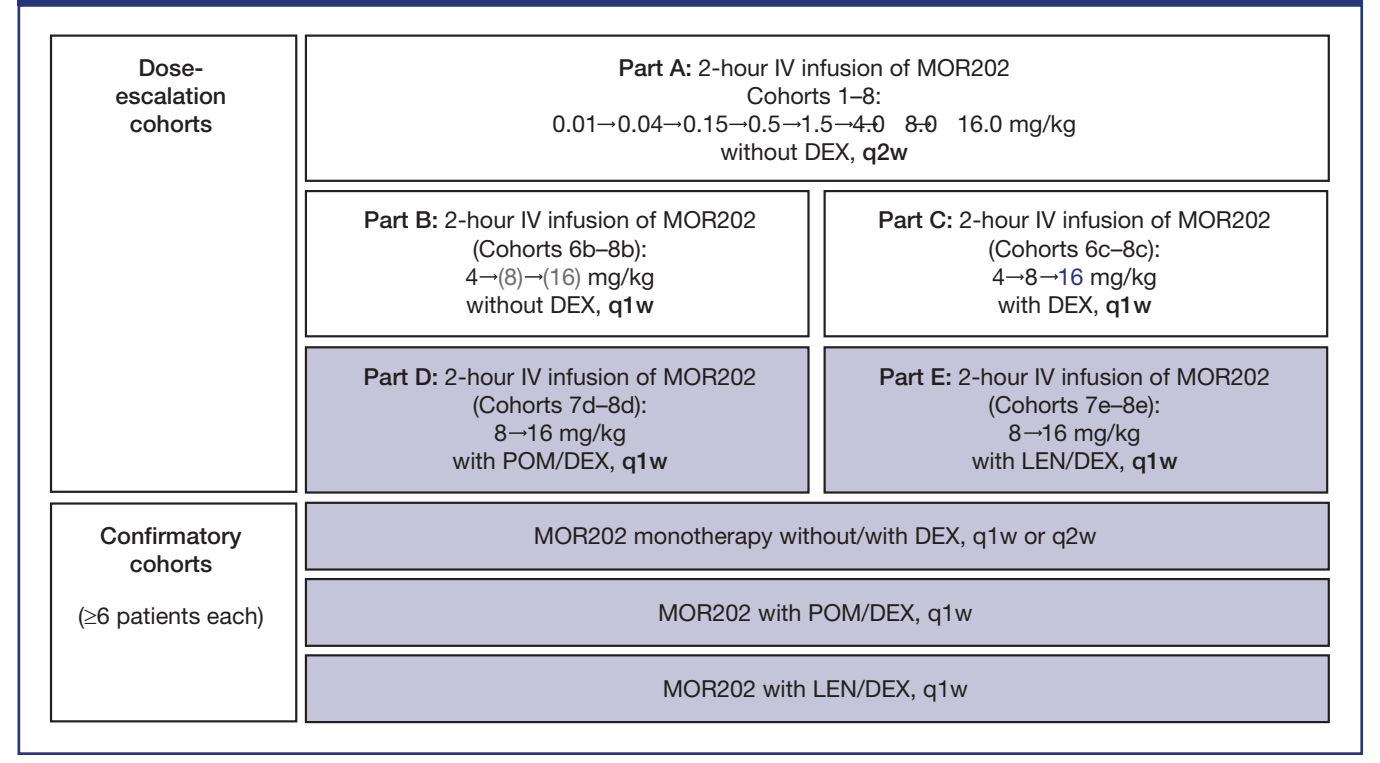
Dose-escalation cohorts
Part A: 2-hour IV infusion of MOR202 Cohorts 1–8: 0.01→0.04→0.15→0.5→1.5→4.0 8.0 16.0 mg/kg without DEX, q2w
Part B: 2-hour IV infusion of MOR202 (Cohorts 6b–8b): 4→(8)→(16) mg/kg without DEX, q1w
Part C: 2-hour IV infusion of MOR202 (Cohorts 6c–8c): 4→8→16 mg/kg with DEX, q1w
Part D: 2-hour IV infusion of MOR202 (Cohorts 7d–8d): 8→16 mg/kg with POM/DEX, q1w
Part E: 2-hour IV infusion of MOR202 (Cohorts 7e–8e): 8→16 mg/kg with LEN/DEX, q1w
Confirmatory cohorts
(≥6 patients each)
MOR202 monotherapy without/with DEX, q1w or q2w
MOR202 with POM/DEX, q1w
MOR202 with LEN/DEX, q1w

q1w, weekly; q2w, every 2 weeks; DEX, dexamethasone; IV, intravenous; LEN, lenalidomide; POM, pomalidomide. Cohorts of part C (weekly cohort 8c), parts D–E (cohorts 7d/e–8d/e) and the 3 confirmation cohorts are planned (see blue text and area shaded blue). Cohort 7b and 8b (8 and 16 mg/kg w/o DEX) did not take place according to Data Monitoring Committee recommendation. During cycle 1 patients in all cohorts received/will receive a loading dose on day 4.

- Parts A–C were conducted in patients with rrMM who had failed 2 previous therapies including an immunomodulatory drug and a proteasome inhibitor.
- Duration of treatment:
 - Patients from part A (cohorts 1–6) were treated for up to 2 cycles only (2 × 28 days)
 - Patients from subsequent cohorts were/will be treated until disease progression (PD) for up to 2 years.
- Route of MOR202 administration: 2-hour intravenous (IV) infusion.
- Premedication: antipyretic, histamine H1 receptor blocker.
- On completion of parts A–E, confirmatory cohorts (of ≥6 patients each) are planned to validate the MTD and/or recommended dose/schedule of MOR202 as monotherapy with and without DEX (q1w or q2w), and in combination with POM/DEX and LEN/DEX (q1w).

Patients and methods

- **Main inclusion criteria:**
 - Adult patients with rrMM who had failed ≥2 previous therapies, including an immunomodulatory agent and a proteasome inhibitor (parts A–C)
 - Documented progression during or after the last prior therapy for MM
 - Serum M-protein ≥0.5 g/100 mL and/or urine M-protein ≥200 mg/24-hour period; absolute neutrophil count (ANC) ≥1,000/mm³.
- **Main exclusion criteria:** primary refractory MM; solitary plasmacytoma or plasma cell leukemia; previous allogenic stem cell transplant.

Results

- This is the first report of safety, preliminary PK and efficacy data for patients in part A and most patients in parts B and C.
- This study is ongoing and higher doses of MOR202 will be evaluated in patients as monotherapy with and without DEX in parts B–C, with immunomodulatory agents in patients in parts D–E, and in phase IIa confirmatory cohorts (Figure 2).
- As of April 13, 2015, 42 patients had been treated; 14 and 28 patients in cohorts 1–4 and 5–8, respectively (Table 1).

Table 1. Patient baseline characteristics

Cohorts Dosing (mg/kg) Patients treated	1–4 0.01–0.5 14	5–8 1.5–16 28	Total 42
Median age (years)	70	69.5	69.5
Gender, %			
Female	42.9	28.6	33.3
Male	57.1	71.4	66.7
Ethnicity, %			
Caucasian	100	100	100
Median Karnofsky PS, %	90	90	90
Median number of prior therapy lines (range)	4 (2–10)	4 (2–11)	4 (2–11)
ASCTs, %	93	75	81
Prior therapies, % (selection)			
Bortezomib	100	100	100
Lenalidomide	100	96.4	97.6
Melphalan	100	96.4	97.6
Cyclophosphamide	64.3	82.1	76.2
Doxorubicin	57.1	60.7	59.5
Thalidomide	35.7	39.3	38.1
Pomalidomide	0	21.4	14.3
Carfilzomib	0	7.1	4.8

ASCTs, autologous stem cell transplants; PS, performance status.

Preliminary efficacy

Data of patients (cohorts 5–8) receiving ≥1 treatment cycle.

- Time on study and best response (Figure 3).
- Best change in M-protein (Figure 4).

Figure 3. Time on study and best response

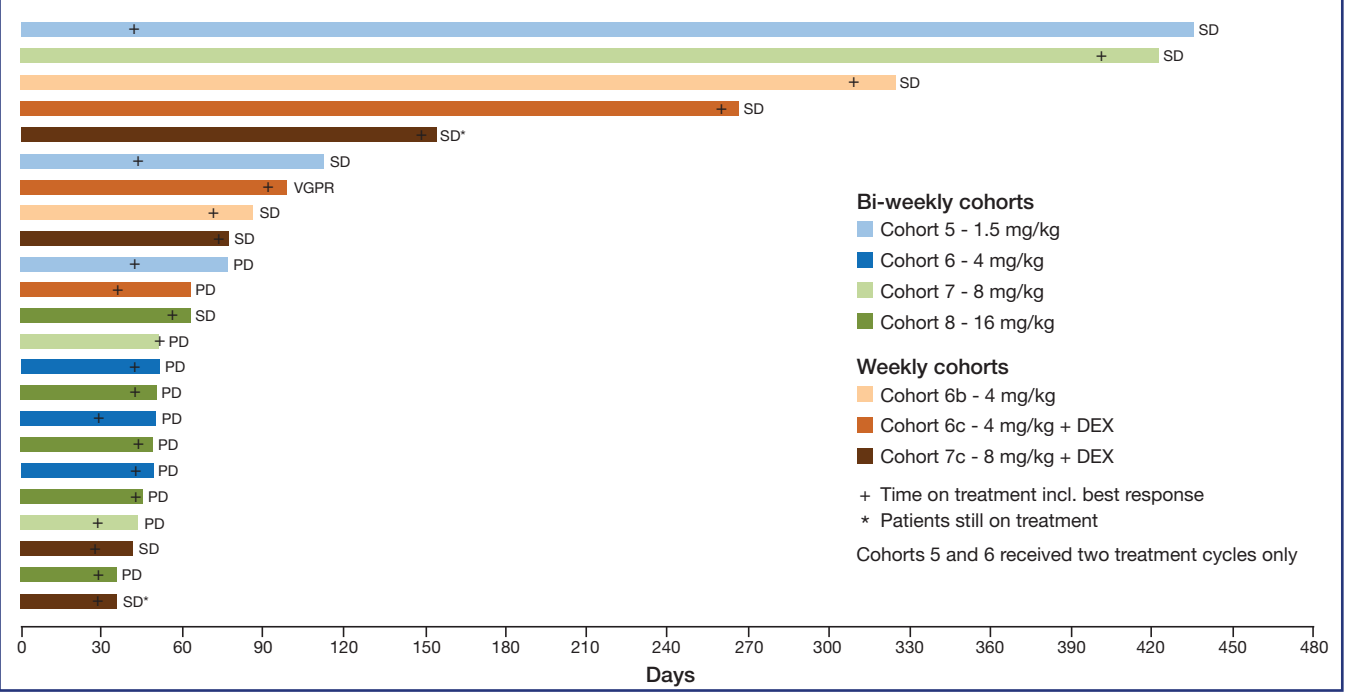
Bi-weekly cohorts
Cohort 5 - 1.5 mg/kg
Cohort 6 - 4 mg/kg
Cohort 7 - 8 mg/kg
Cohort 8 - 16 mg/kg
Weekly cohorts
Cohort 6b - 4 mg/kg
Cohort 6c - 4 mg/kg + DEX
Cohort 7c - 8 mg/kg + DEX
+ Time on treatment incl. best response
* Patients still on treatment
Cohorts 5 and 6 received two treatment cycles only
SD
VGPR
PD
SD*
Days
0
30
60
90
120
150
180
210
240
270
300
330
360
390
420
450
480

DEX, dexamethasone; PD, progressive disease; SD, stable disease; VGPR, very good partial response.

Figure 4. Best change in M-protein

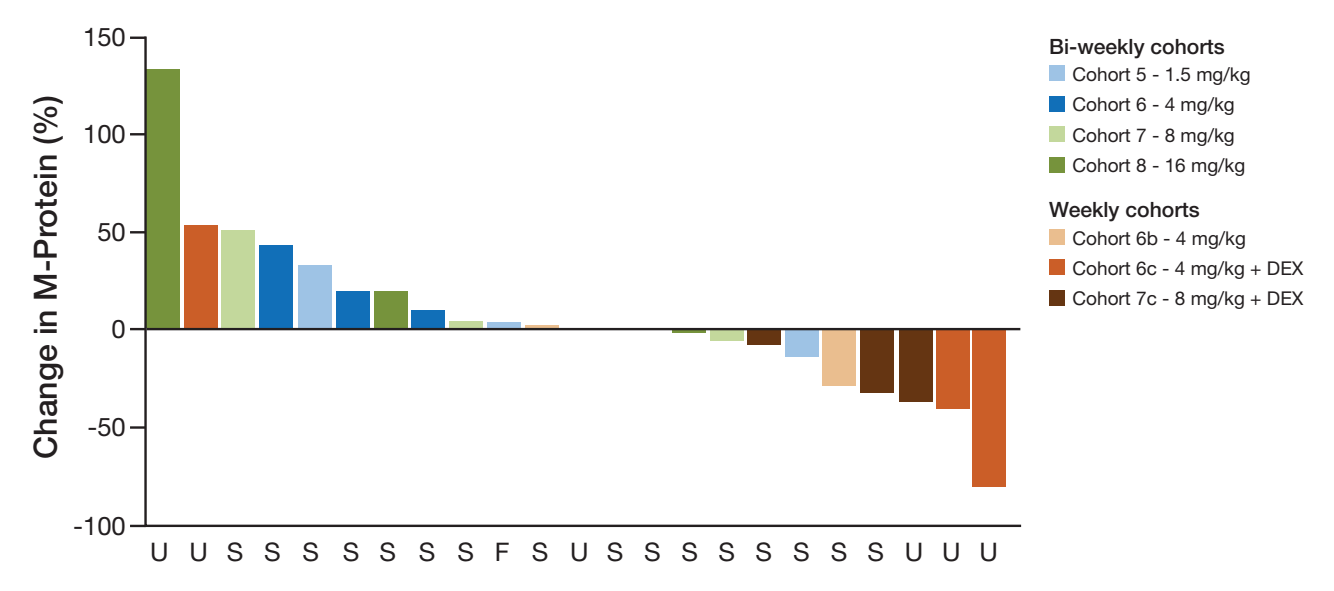
Change in M-Protein (%)
150
100
50
0
-50
-100
U U S S S S S S S F S U S S S S S S S S U U U
Bi-weekly cohorts
Cohort 5 - 1.5 mg/kg
Cohort 6 - 4 mg/kg
Cohort 7 - 8 mg/kg
Cohort 8 - 16 mg/kg
Weekly cohorts
Cohort 6b - 4 mg/kg
Cohort 6c - 4 mg/kg + DEX
Cohort 7c - 8 mg/kg + DEX

DEX, dexamethasone; F, free light chain kappa [FLCk]; S, serum; U, urine.

Safety

- 42 (100%) patients experienced AEs during treatment irrespective of causality.
- 18 (42.9%) patients discontinued treatment due to AEs, in 3 (7.1%) cases due to a suspected causal relationship.
- There have been no treatment-related deaths.
- The MTD of MOR202 has not yet been reached.

Table 2. Most frequently reported AEs

Cohorts	1–4		5–8		Total	
Grades	≥3	All	≥3	All	≥3	All
Hematologic AEs* (≥10%)						
Anemia	7.1	42.9	-	28.6	2.4	33.3
Lymphocyte count decreased	-	7.1	21.4	25.0	14.3	19.0
WBC count decreased	7.1	7.1	7.1	25.0	7.1	19.0
Leukopenia	-	7.1	7.1	17.9	4.8	14.3
Neutrophil count decreased	-	0.0	3.6	17.9	2.4	11.9
Thrombocytopenia	-	0.0	7.1	17.9	4.8	11.9
Non-hematologic AEs (≥15%)						
Fatigue	-	42.9	-	28.6	-	33.3
Nausea	-	35.7	-	21.4	-	26.2
Diarrhea	-	28.6	-	14.3	-	19.0
Headache	-	42.9	-	3.6	-	16.7
Nasopharyngitis	-	28.6	-	10.7	-	16.7
Pyrexia	-	21.4	-	14.3	-	16.7

*MedDRA System Organ Classes "Blood and Lymphatic System Disorders" and "Investigations" are applicable and therefore Preferred Terms from both are included; AEs, adverse events; WBC, white blood cell.

Infusion tolerability

- A 2-hour IV infusion was feasible in all patients.
- Infusion-related reactions (IRRs) occurred in 13 (31%) patients and only in patients receiving MOR202 without DEX, mainly during the first infusion.
- All IRRs were grade 1–2 except for 1 patient (grade 3).
- No IRRs occurred in patients who received DEX.

Figure 5. Infusion-related reactions

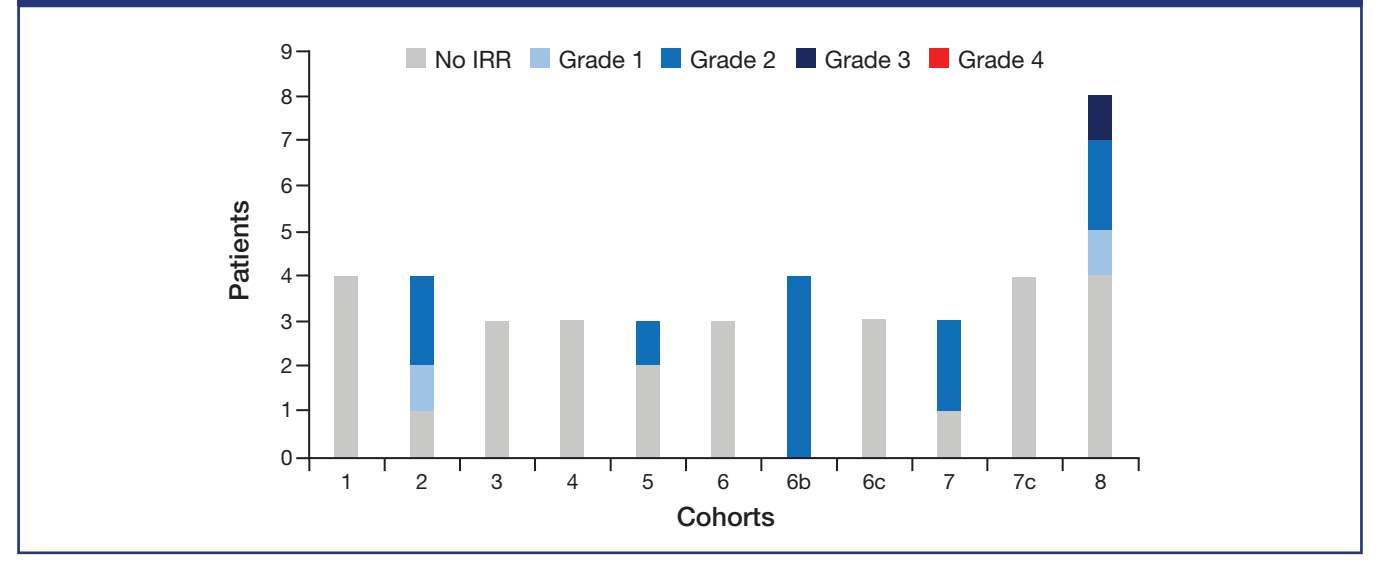
No IRR
Grade 1
Grade 2
Grade 3
Grade 4
Patients
0
1
2
3
4
5
6
7
8
9
1
2
3
4
5
6
6b
6c
7
7c
8
Cohorts

Pharmacokinetics and immunogenicity

- In most patients treated q2w, a dominant target-mediated sink effect was observed leading to low or no detectable serum trough levels (Figure 6A, ≥4 mg/kg only).
- In contrast, patients treated q1w (≥4 mg/kg) showed constant or slightly accumulating trough levels (Figure 6B).
- Data indicate that further dose-escalation might lead to full target saturation at higher dose levels.
- A terminal elimination half-life of 2–3 weeks was estimated by comparing modelled serum concentrations of MOR202 (at 4 mg/kg, q1w) with measured patient data.
- Only 1 patient (0.15 mg/kg, q2w) developed a transient anti-MOR202 antibody response.

Figure 6. MOR202 serum concentrations over time for patients dosed at ≥4mg/kg. Initial two cycles following: A. q2w dosing; B. q1w dosing.

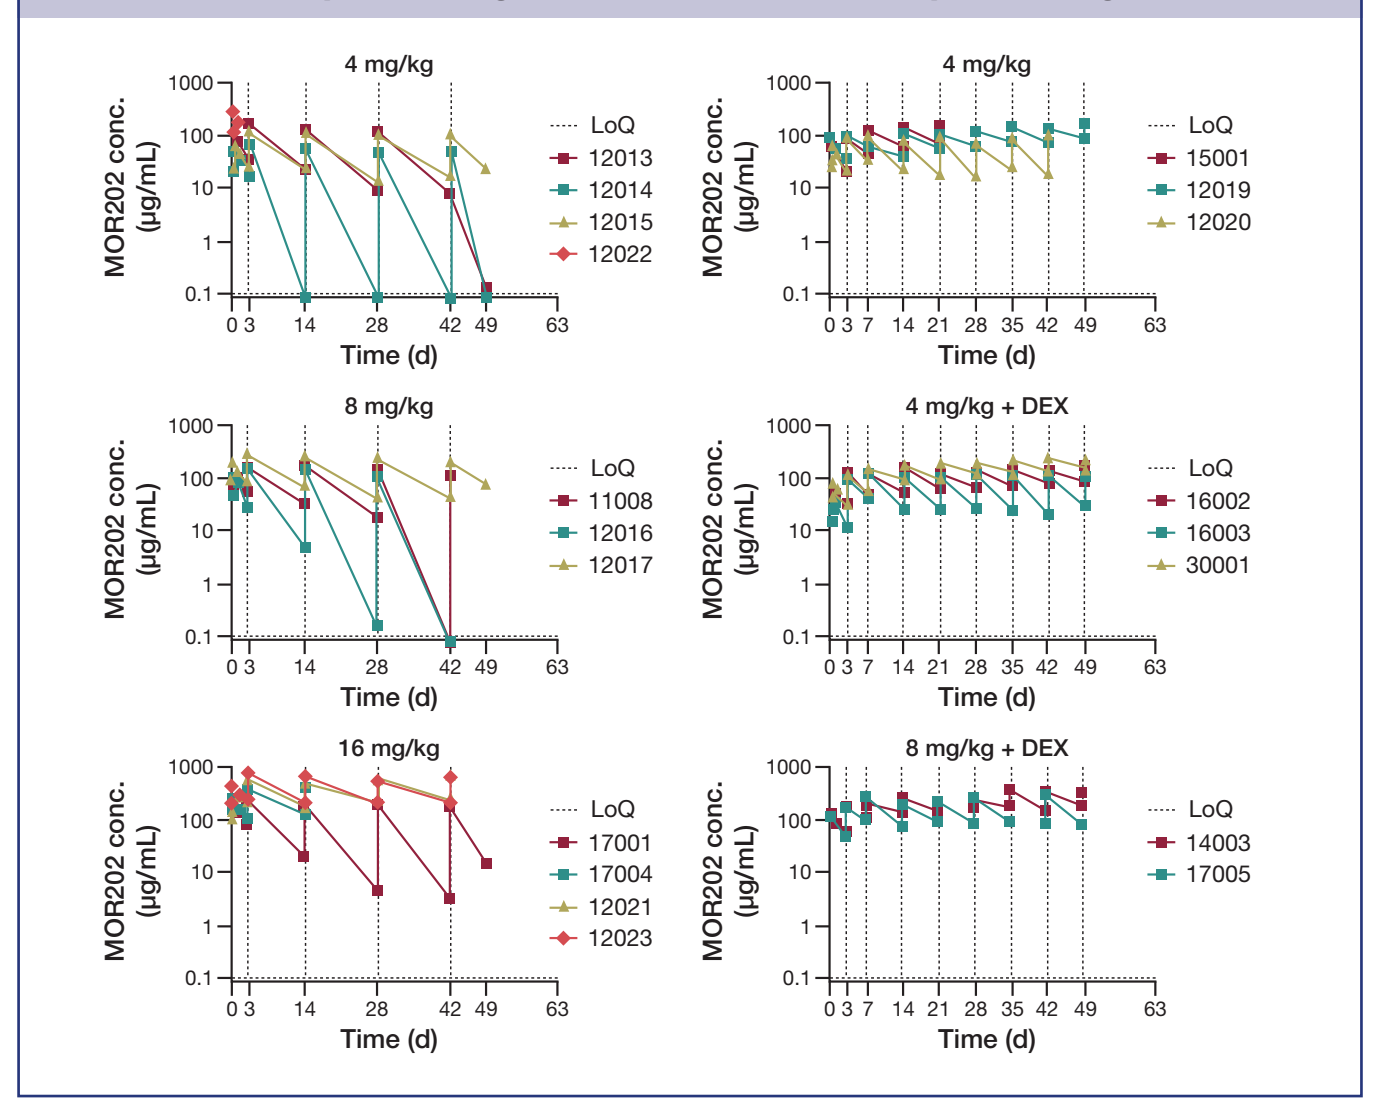
A. q2w dosing
B. q1w dosing
4 mg/kg
LoQ
12013
12014
12015
12022
4 mg/kg
LoQ
15001
12019
12020
8 mg/kg
LoQ
11008
12016
12017
4 mg/kg + DEX
LoQ
16002
16003
30001
16 mg/kg
LoQ
17001
17004
12021
12023
8 mg/kg + DEX
LoQ
14003
17005
MOR202 conc. (µg/mL)
1000
100
10
1
0.1
Time (d)
0 3 14 28 42 49 63
0 3 7 14 21 28 35 42 49 63

q1w, weekly; q2w, every two weeks; LoQ, limit of quantification. Both dosing regimens included a loading dose on day 4.

Case report of a very good partial response (VGPR)

- Female, 45 years of age, Karnofsky PS 90%.
- First diagnosis: February 2006.
- MM staging: International staging system III; Salmon and Durie IIIB.
- 4 prior therapies, including an autologous stem cell transplant.
- Chromosomal aberration: del13.
- Allocated to part C, cohort 6 (4 mg/kg, q1w, with DEX).
- 15 infusions received.
- VGPR was determined after 29 days on treatment.
- The patient developed new bone lesions after 84 days on treatment.

Conclusions

- The MTD of MOR202 has not yet been reached.
- MOR202 can be safely administered as a 2-hour IV infusion and is well tolerated.
- In the cohorts without DEX the incidence of IRRs was low and mainly limited to the first infusion.
- No IRRs were reported in patients treated with MOR202 plus DEX.
- In this heavily pre-treated rrMM population, encouraging long-lasting tumor control has been observed already at early cohorts.
- PK data indicate the potential for full target occupancy in patients who receive 16 mg/kg MOR202, q1w.
- Further cohorts will evaluate doses of MOR202 up to 16 mg/kg q1w with DEX, as well as in combination with POM/DEX and LEN/DEX.

References

1. Lin, et al. Am J Clin Pathol 2004; 121:482–8.
2. Angelopoulou, et al. Eur J Haematol 2002; 68:12–21.
3. Schwonzen, et al. Br J Haematol 1993; 83:232–9.
4. Keyhani, et al. Leukemia Res 1999; 24:153–9.
5. Domingo-Domènech, et al. Haematologica 2002; 87:1021–7.
6. Boxhammer, et al. Proceedings of the 54th ASH annual meeting 2012; abstr 4018.
7. Endell, et al. J Clin Oncol 2011; 29 (suppl; abstr 8078).

Acknowledgements

This study was sponsored by MorphoSys AG. Medical writing support was provided by Dr Matthew Young, Cancer Communications and Consultancy Ltd, Knutsford, UK, and funded by MorphoSys AG.

Copies of this poster obtained through Quick Response code (QR) are for personal use only and may not be reproduced without the permission from ASCO and the authors of this poster